Adaptive transcriptional and immunomodulatory responses to the novel microtubule destabilizer AUS_001 in pancreatic and colorectal cancer models

FPN: 4879eP

Marina Koutsioumpa[1], Herman Lelie[1], Lina Kuo[1]

[1] Australis Pharmaceuticals, Inc., Simi Valley, CA, USA

Background

Heterogeneous and therapy-resistant gastrointestinal cancers continue to pose major clinical challenges, calling for novel treatment strategies. Our prior studies revealed that the prenylated hydroxy-stilbene, AUS_001, inhibits tubulin polymerization via its unique binding configuration to the colchicine site of β-tubulin and suppresses cancer cell proliferation both *in vitro* and *in vivo*.

Aim

The goal of the current study was to perform long-term *in vitro* studies that mimic chronic therapy, allowing more predictive insights regarding side effects, chemoresistance and rational drug combinations.

Results

1. CAPAN-1 cells exhibit adaptive resistance upon long-term AUS_001 exposure

Figure 1. (A) Methodology for the establishment of AUS_001 Resistant CAPAN-1 pancreatic cancer cells. The IC50 of the parental cells equals 2.67 μM. **(B)** EVOS™ XL Core Imaging System of CAPAN-1 cells with acquired AUS_001 resistance after 90 days in culture. **(C)** Cell viability assessment and **(D)** semi-quantitative detection of poly (ADP-ribose) deposited by PARP-1 onto immobilized histone proteins in lysates originating from parental (wt) or AUS_001-Resistant CAPAN-1 cells treated with 20μM AUS_001 and the respective vehicle for 72 hours.

1A

CAPAN_1 cells treated with 2.5 uM AUS_001 every 3 days

↓

Discard apoptotic cells and amplify surviving cells with AUS_001-containing medium for 90 days

↓

Induce process with double AUS_001 concentration (Repeat 3x)

↓

20_uM AUS_001 Resistant CAPAN_1 cells

↓

After 90 days in culture under the presence of 20uM AUS_001, acquired resistance of CAPAN_1 cells to AUS_001 was assessed by :

Cell Viability upon 3 days of treatment with 20uM AUS_001 | Poly (ADP-ribose) polymerase activation status

1B

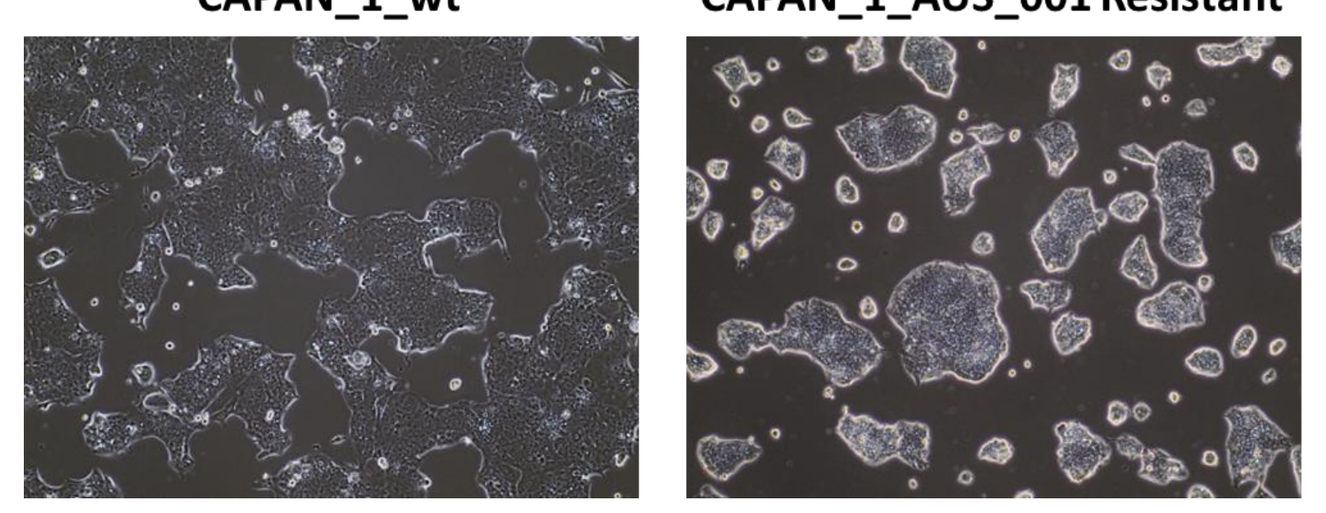

CAPAN_1_wt
CAPAN_1_AUS_001 Resistant

1C

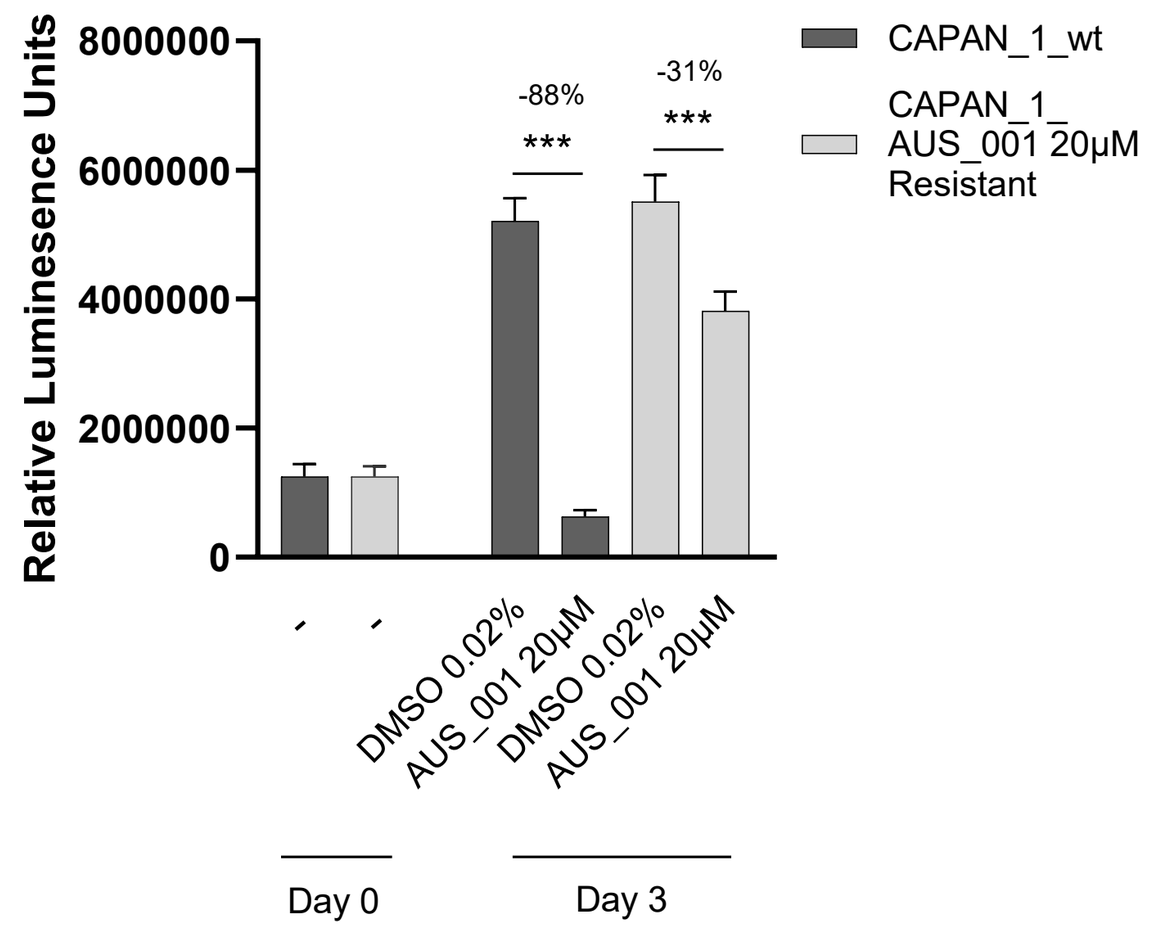

Relative Luminesence Units
8000000
6000000
4000000
2000000
0
-88%

-31%

CAPAN_1_wt
CAPAN_1_ AUS_001 20μM Resistant
DMSO 0.02%
AUS_001 20μM
DMSO 0.02%
AUS_001 20μM
Day 0
Day 3

1D

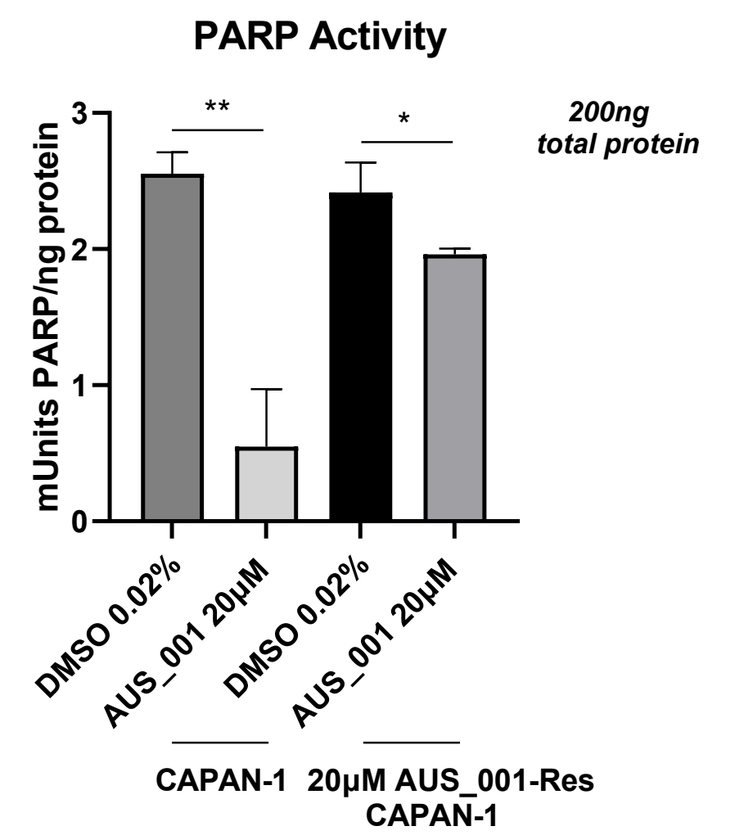

PARP Activity
mUnits PARP/ng protein
200ng total protein
**
*
DMSO 0.02%
AUS_001 20μM
DMSO 0.02%
AUS_001 20μM
CAPAN-1
20μM AUS_001-Res CAPAN-1

2. Gene ontology analysis for DEGs in signaling pathways and molecular processes under prolonged AUS_001 pressure

2A

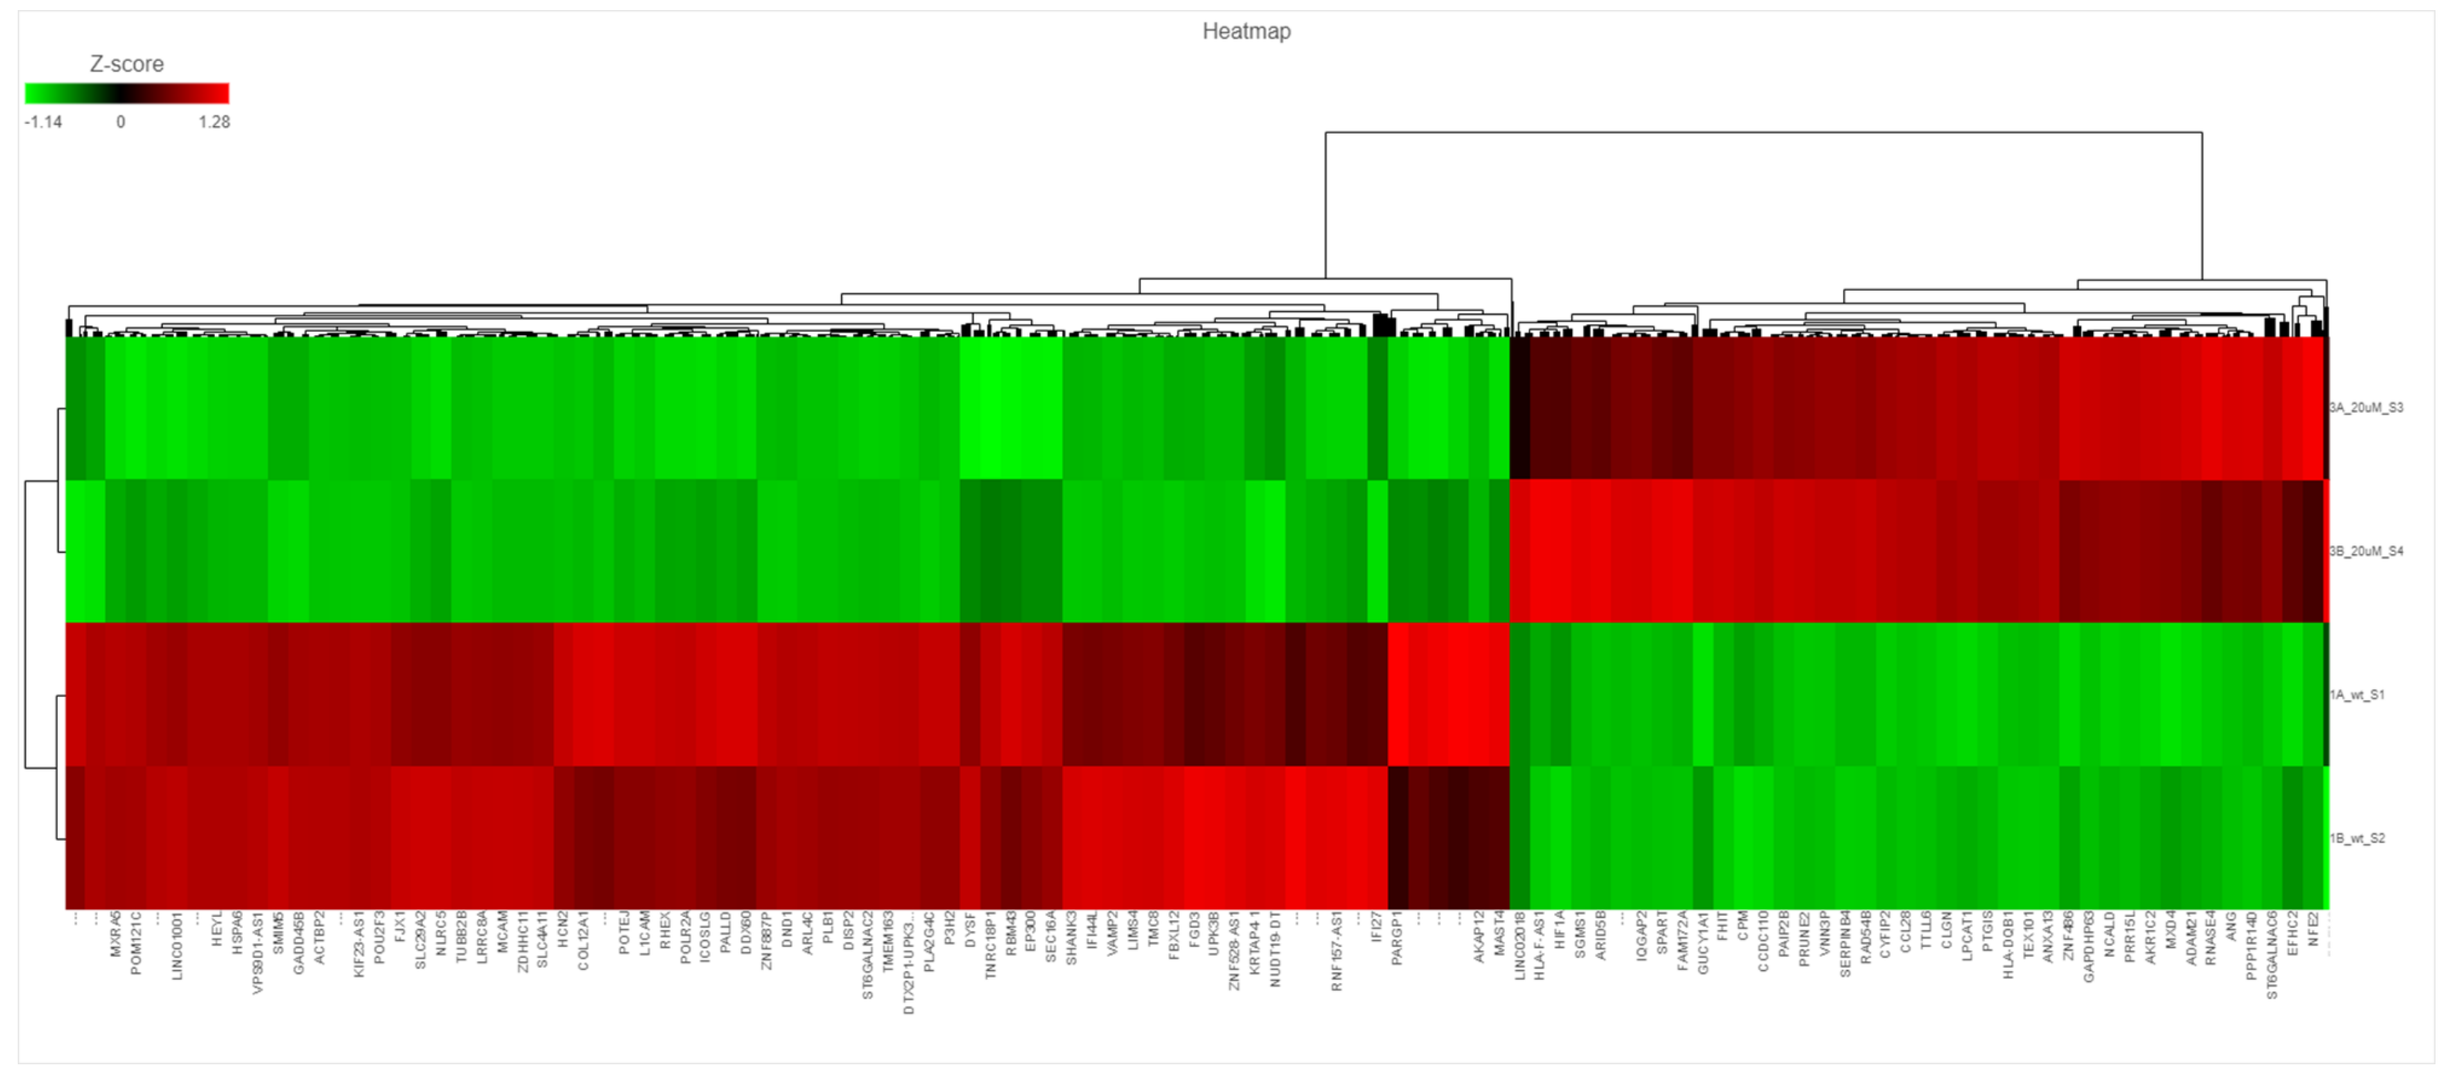

Heatmap
Z-score

2B

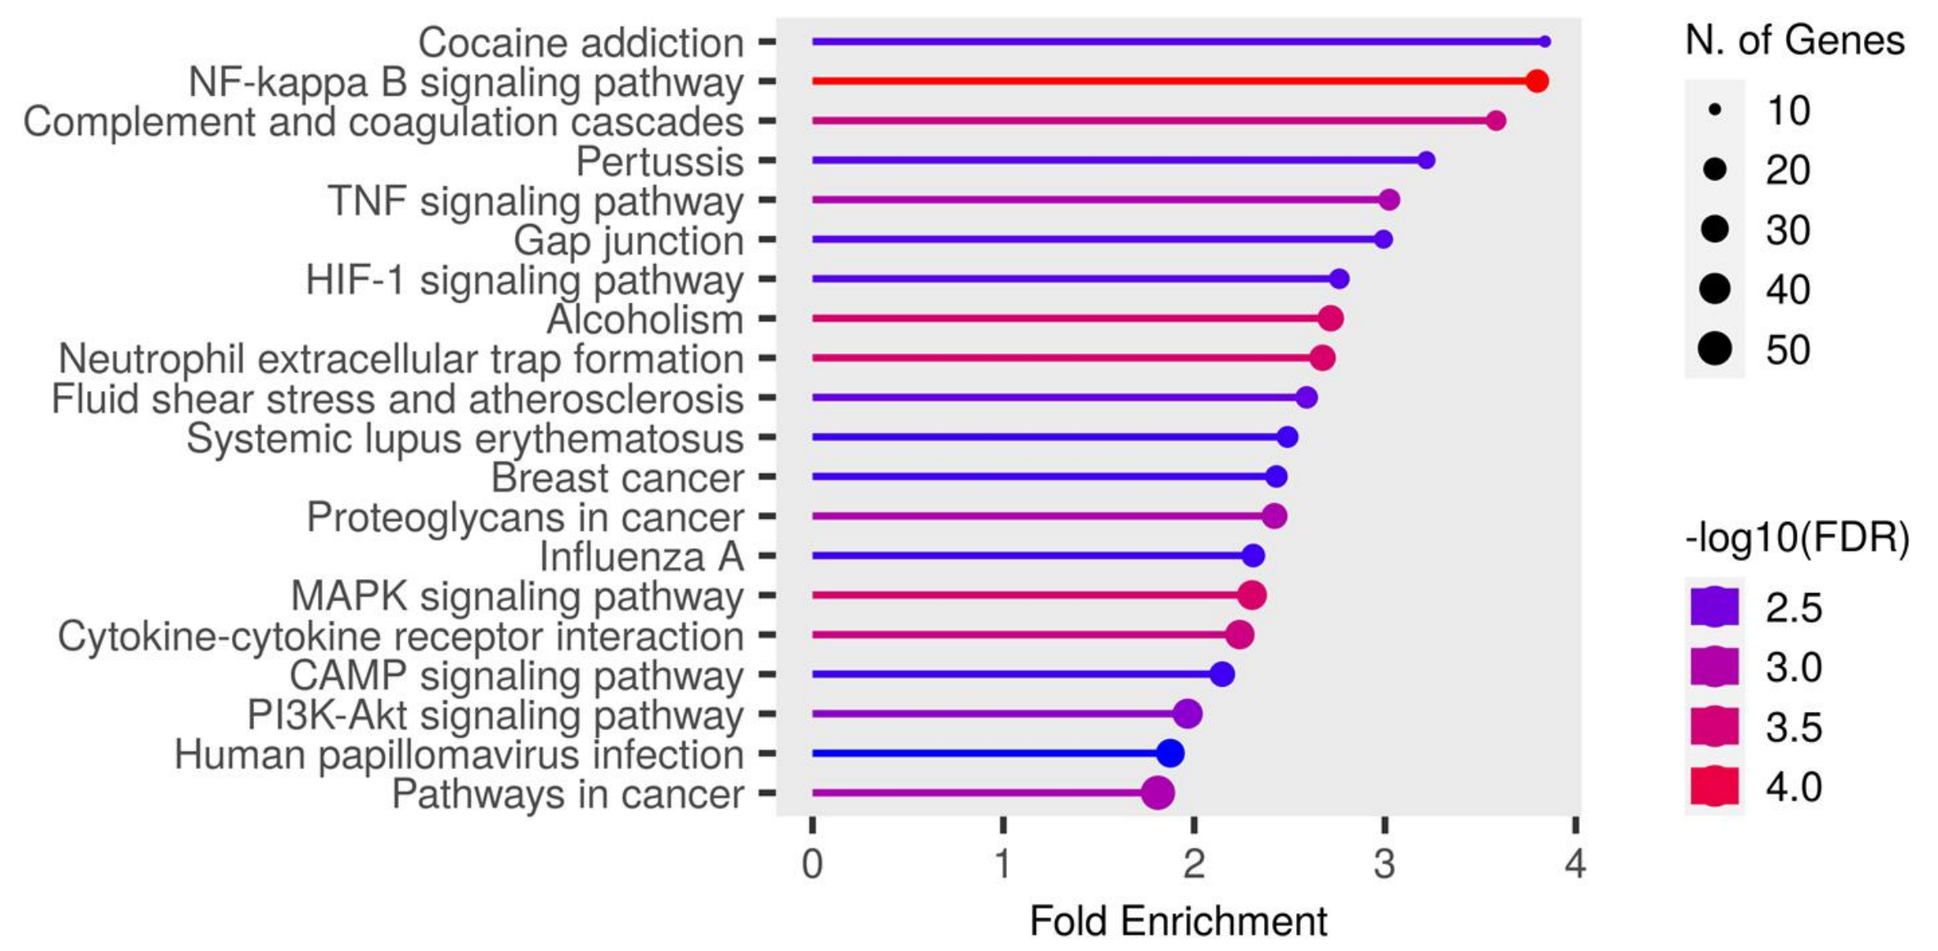

Cocaine addiction
NF-kappa B signaling pathway
Complement and coagulation cascades
Pertussis
TNF signaling pathway
Gap junction
HIF-1 signaling pathway
Alcoholism
Neutrophil extracellular trap formation
Fluid shear stress and atherosclerosis
Systemic lupus erythematosus
Breast cancer
Proteoglycans in cancer
Influenza A
MAPK signaling pathway
Cytokine-cytokine receptor interaction
CAMP signaling pathway
PI3K-Akt signaling pathway
Human papillomavirus infection
Pathways in cancer
Fold Enrichment
N. of Genes
-log10(FDR)

2C

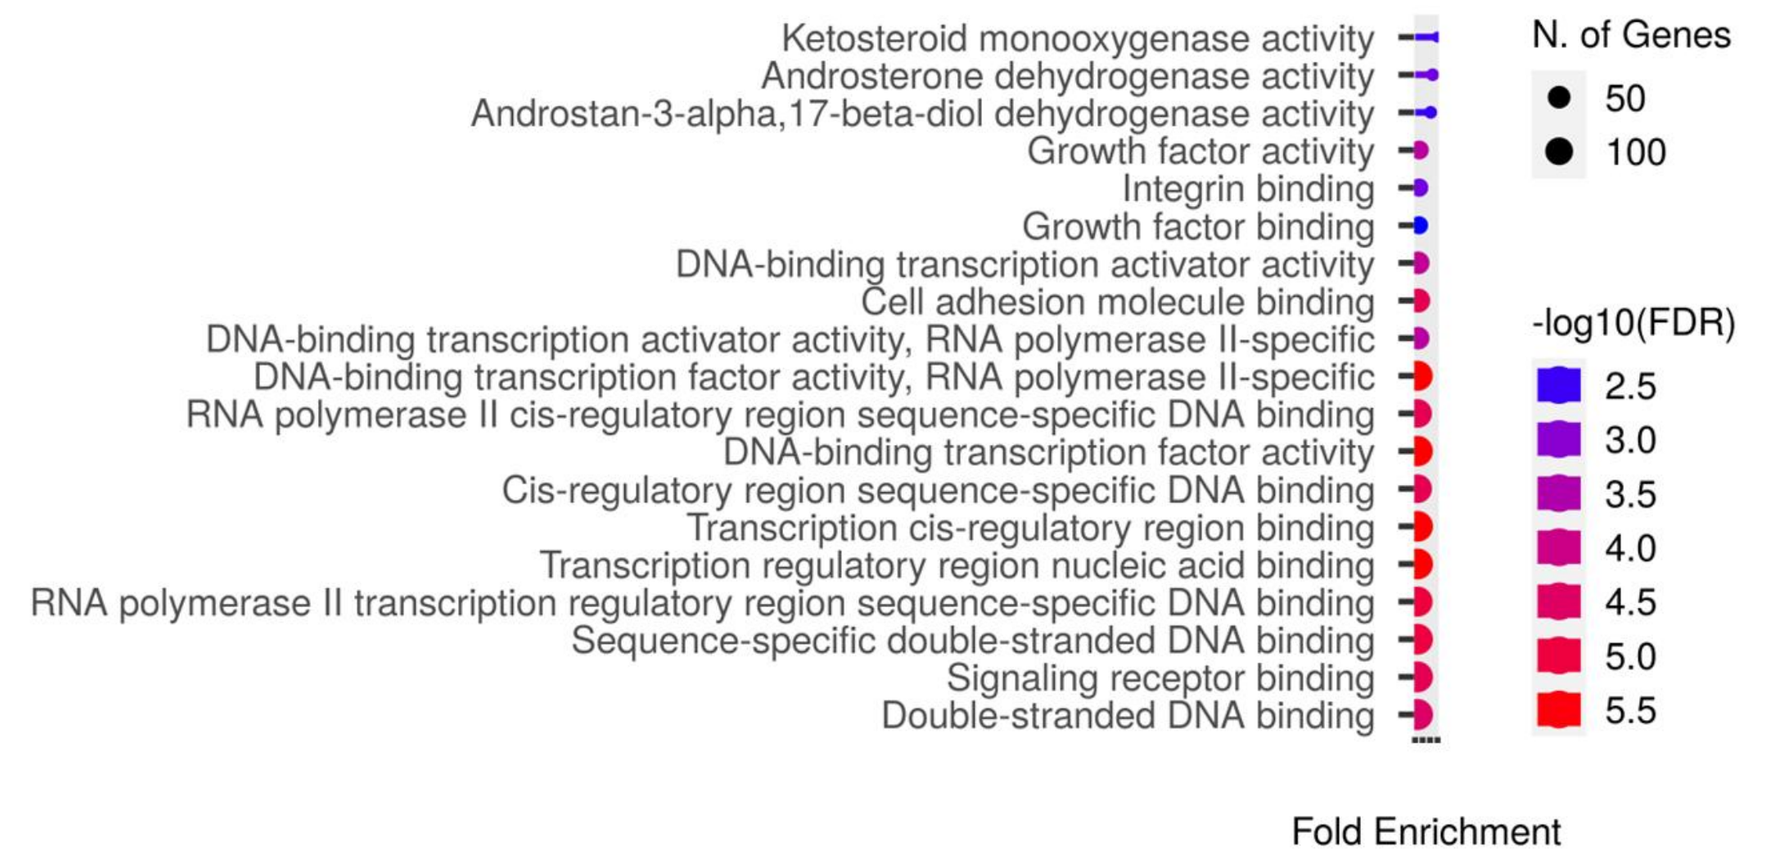

Ketosteroid monooxygenase activity
Androsterone dehydrogenase activity
Androstan-3-alpha,17-beta-diol dehydrogenase activity
Growth factor activity
Integrin binding
Growth factor binding
DNA-binding transcription activator activity
Cell adhesion molecule binding
DNA-binding transcription activator activity, RNA polymerase II-specific
DNA-binding transcription factor activity, RNA polymerase II-specific
RNA polymerase II cis-regulatory region sequence-specific DNA binding
DNA-binding transcription factor activity
Cis-regulatory region sequence-specific DNA binding
Transcription cis-regulatory region binding
Transcription regulatory region nucleic acid binding
RNA polymerase II transcription regulatory region sequence-specific DNA binding
Sequence-specific double-stranded DNA binding
Signaling receptor binding
Double-stranded DNA binding
Fold Enrichment
N. of Genes
-log10(FDR)

Figure 2. (A) Heatmap with hierarchical clustering of RNA Sequencing–derived differentially expressed genes (DEGs) between parental (wt) and AUS_001 Resistant CAPAN-1 cells. The data is displayed in a grid where each row represents a gene and each column represents a sample. The color and intensity of the boxes is used to represent changes (not absolute values) of gene expression. DEGs are clustered within **(B)** functional categories, including signaling pathways that play critical roles in cellular response to external stimuli, and **(C)** molecular functions linked to transcriptional control.

3. Gene signature of pancreatic cancer cells with acquired AUS_001 resistance

3A

Gene name	Gene Description	P-value	Fold change
TUBB2A	tubulin beta 2A class IIa [Source:HGNC Symbol;Acc:HGNC:12412]	0.000000257	-4.41
TUBB2B	tubulin beta 2B class IIb [Source:HGNC Symbol;Acc:HGNC:30829]	0.00000155	-3.83
TUBB3	tubulin beta 3 class III [Source:HGNC Symbol;Acc:HGNC:20772]	0.00000682	-2.91
TUBB4A	tubulin beta 4A class IVa [Source:HGNC Symbol;Acc:HGNC:20774]	0.000084	-2.38
TUBD1	tubulin delta 1 [Source:HGNC Symbol;Acc:HGNC:16811]	0.0000237	2.2
MAP10	microtubule associated protein 10 [Source:HGNC Symbol;Acc:HGNC:29265]	2.72E-04	-2.01
MAP9	microtubule associated protein 9 [Source:HGNC Symbol;Acc:HGNC:26118]	4.35E-05	2.22E+00
MAPT	microtubule associated protein tau [Source:HGNC Symbol;Acc:HGNC:6893]	1.47E-06	3.17
MAP2	microtubule associated protein 2 [Source:HGNC Symbol;Acc:HGNC:6839]	0.0000558	3.38
JAKMIP3	Janus kinase and microtubule interacting protein 3 [Source:HGNC Symbol;Acc:HGNC:23523]	0.00381	2.22
TTBK2	tau tubulin kinase 2 [Source:HGNC Symbol;Acc:HGNC:19141]	0.0000345	2.23
MACF1	microtubule actin crosslinking factor 1 [Source:HGNC Symbol;Acc:HGNC:13664]	0.000284	-2.03
MICAL3	microtubule associated monooxygenase, calponin and LIM domain containing 3 [Source:HGNC Symbol;Acc:HGNC:24694]	0.000738	-2.1
MAST4	microtubule associated serine/threonine kinase family member 4 [Source:HGNC Symbol;Acc:HGNC:19037]	0.000133	-2.2
TTLL6	tubulin tyrosine ligase like 6 [Source:HGNC Symbol;Acc:HGNC:26664]	0.000017	2.48
TPPP3	tubulin polymerization promoting protein family member 3 [Source:HGNC Symbol;Acc:HGNC:24162]	0.0000182	3.13
ATAT1	alpha tubulin acetyltransferase 1 [Source:HGNC Symbol;Acc:HGNC:21186]	0.00011	-2.09
ACTB	actin beta [Source:HGNC Symbol;Acc:HGNC:132]	0.0000034	-2.37
DNAH12	dynein axonemal heavy chain 12 [Source:HGNC Symbol;Acc:HGNC:2943]	0.0000211	-2.44
DYNC2H1	dynein cytoplasmic 2 heavy chain 1 [Source:HGNC Symbol;Acc:HGNC:2962]	0.000591	2.04
DNAH6	dynein axonemal heavy chain 6 [Source:HGNC Symbol;Acc:HGNC:2951]	0.000159	2.47
KIF5C	kinesin family member 5C [Source:HGNC Symbol;Acc:HGNC:6325]	0.0000159	6.03
KIF12	kinesin family member 12 [Source:HGNC Symbol;Acc:HGNC:21495]	0.000914	2.29

3B

Gene name	Gene Description	P-value	Fold change
HLA-DMA	major histocompatibility complex, class II, DM alpha [Source:HGNC Symbol;Acc:HGNC:4934]	1.27E-05	2.46
CIITA	class II major histocompatibility complex transactivator [Source:HGNC Symbol;Acc:HGNC:7067]	3.87E-06	3.6
HLA-DPB1	major histocompatibility complex, class II, DP beta 1 [Source:HGNC Symbol;Acc:HGNC:4940]	0.0000261	4.24
HLA-DMB	major histocompatibility complex, class II, DM beta [Source:HGNC Symbol;Acc:HGNC:4935]	0.000000796	5.49
HLA-DPA1	major histocompatibility complex, class II, DP alpha 1 [Source:HGNC Symbol;Acc:HGNC:4938]	0.00000111	9.11
HLA-DQB1	major histocompatibility complex, class II, DQ beta 1 [Source:HGNC Symbol;Acc:HGNC:4944]	0.000000329	12.7
HLA-DRB6	major histocompatibility complex, class II, DR beta 6 (pseudogene) [Source:HGNC Symbol;Acc:HGNC:4954]	4.77E-08	12.8
HLA-DRA	major histocompatibility complex, class II, DR alpha [Source:HGNC Symbol;Acc:HGNC:4947]	3.75E-09	14
HLA-DRB1	major histocompatibility complex, class II, DR beta 1 [Source:HGNC Symbol;Acc:HGNC:4948]	2.46E-08	16.8
HLA-DRB5	major histocompatibility complex, class II, DR beta 5 [Source:HGNC Symbol;Acc:HGNC:4953]	0.000000138	20.8

Figure 3. (A) Key alterations in genes associated with microtubule cytoskeleton, cytokinesis and cytoskeleton/microtubule interactions were triggered by AUS_001 sustained exposure. **(B)** Dramatic increase in the expression major histocompatibility complex class II genes was observed under AUS_001 pressure.

4. Oral administration of AUS_001 triggers tumor infiltration of lymphocytes *in vivo*

4A

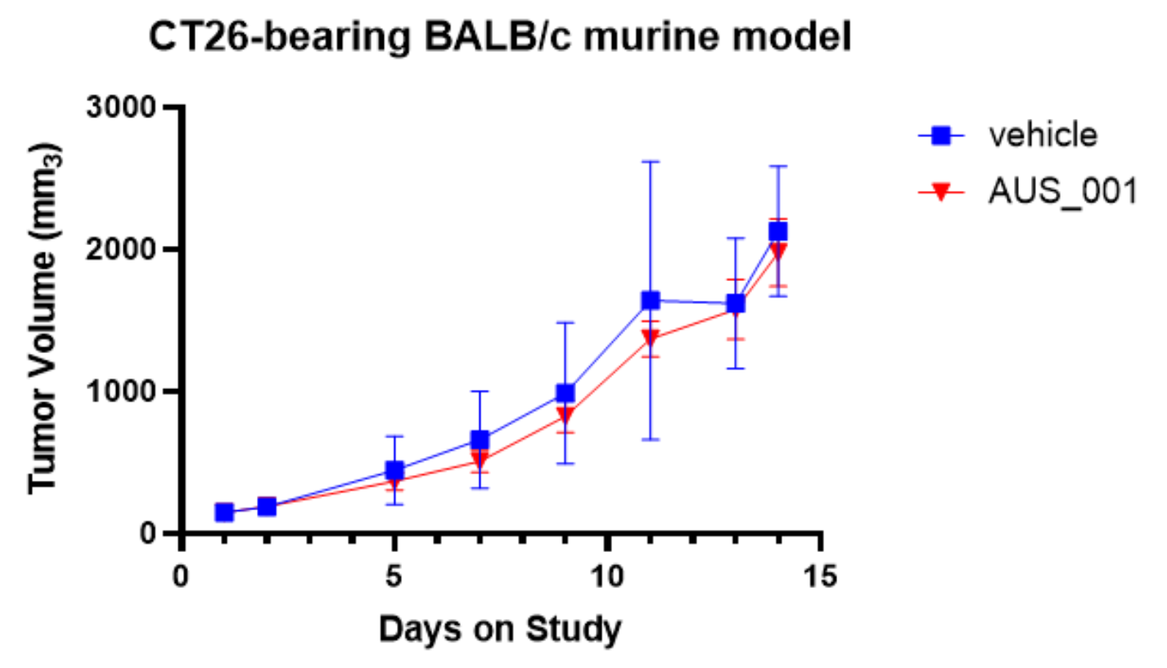

CT26-bearing BALB/c murine model
Tumor Volume (mm3)
Days on Study
vehicle
AUS_001

4B

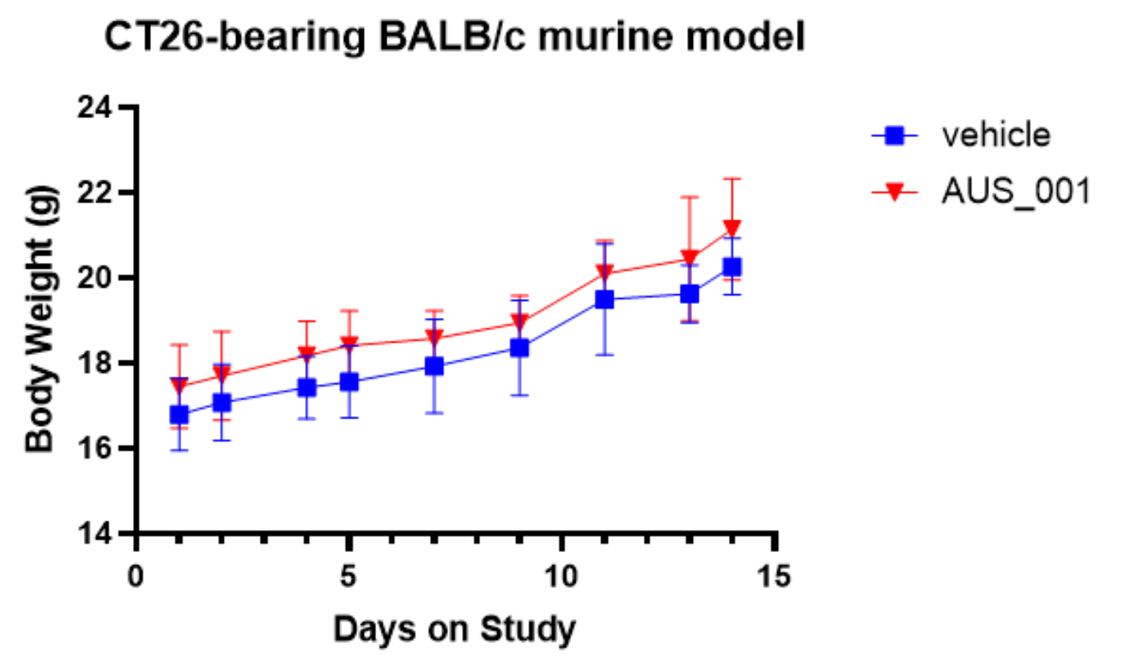

CT26-bearing BALB/c murine model
Body Weight (g)
Days on Study
vehicle
AUS_001

4C

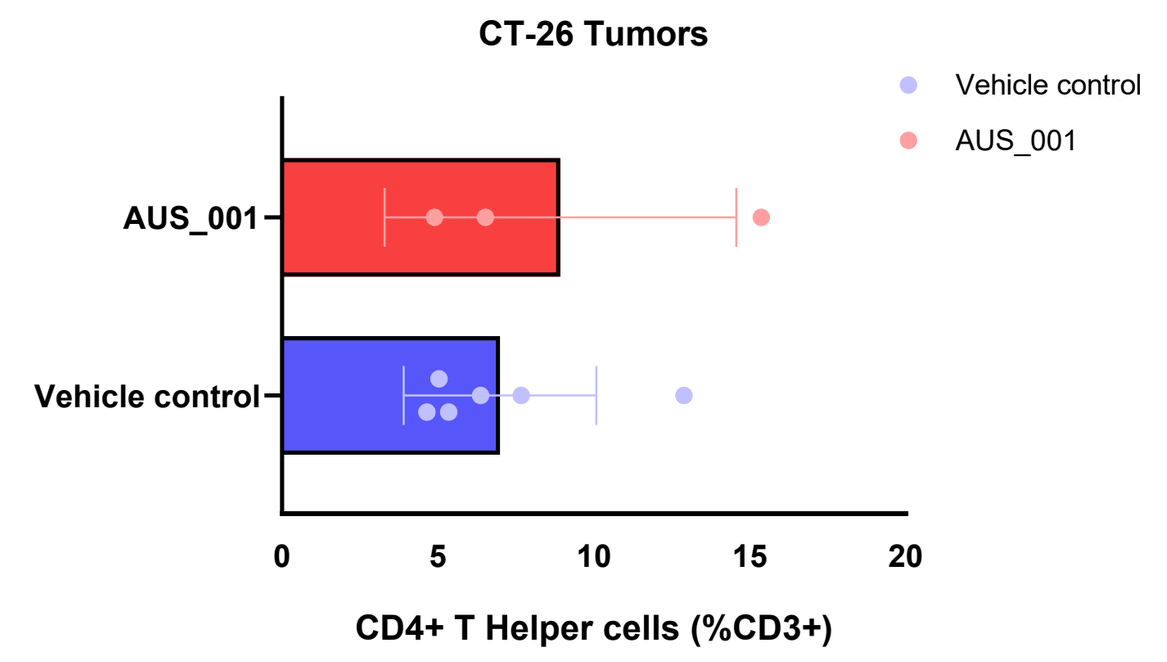

CT-26 Tumors
Vehicle control
AUS_001
CD4+ T Helper cells (%CD3+)

4D

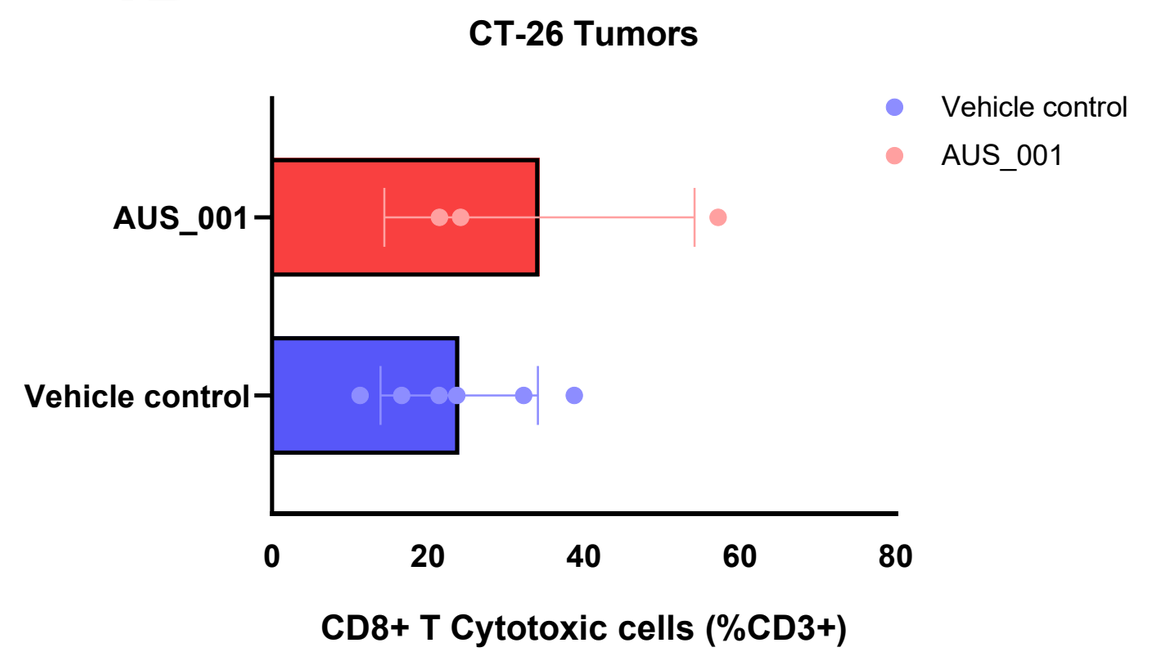

CT-26 Tumors
Vehicle control
AUS_001
CD8+ T Cytotoxic cells (%CD3+)

Figure 4. Assessment of **(A)** tumor volume (mm^3), **(B)** body weight, intra-tumoral infiltration by **(C)** CD4+ T Helper cells and **(D)** CD8+ cytotoxic T cells, following daily oral administration of SEDDS-formulated AUS_001. CT26-bearing syngeneic BALB/c mice (n=5) received 900 mg/kg AUS_001 once daily for 4 days, followed by 450 mg/kg daily for an additional 10 days prior to sacrifice. Freshly excised tumors were enzymatically dissociated and analyzed by flow cytometry.

Conclusions

***Dose-escalation exposure** to AUS-001 drives transcriptional adaptations characterized by cytoskeletal remodeling and immune modulation across pancreatic and colorectal cancer models. Collectively, these findings provide mechanistic insight into AUS-001 activity and support its further investigation as a potential therapeutic strategy for gastrointestinal cancers.*

Email Contact: mkoutsioumpa@australispharma.com

Disclosure: The first and presenting author declares no conflicts of interest.